• Jan-Mar 2015;8(1):2-11. doi: 10.1016/j.optom.2014.12.001. Epub 2015 Jan 7.

JENVIS Vision Research Jena

Ernst-Abbe-Hochschule Jena
University of Applied Sciences

Change in objective ocular redness and symptoms after refit of weekly and monthly lenses into a water-gradient daily disposable lens material

Authors: Sebastian Marx[1], Stefan Schwarz[2], Katharina Keller[1], Wolfgang Sickenberger[1],
Affiliations: [1]JENVIS Research c/o Ernst Abbe University Jena, Germany;
[2]Optometrie Schwarz, Hildesheim, Germany

Introduction:

Healthy, white eyes are naturally desirable for most contact lens wearers. Self-perceived redness is often the only criterion by which wearers perceive their lenses as being suitable and their eyes “healthy” or not. Strategies and products aimed specifically at reducing ocular redness may therefore benefit ECPs.
Contact lens-related hyperemia can be driven by many factors, including limited oxygen flux to the ocular surface, interpalpebral friction increase, tear film layer disruption, pH level changes and others [1]. Diurnal lens dehydration can steepen the lens curvature, reducing lens movement and tear film exchange, inducing conjunctival pressure marks, redness and ocular surface staining [2].
Some lens care solution ingredients can also contribute to hyperemia, through adhesion to the lens material and release on eye. Certain lens types or solutions (sometimes in combination) may be more prone to increasing hyperemia than others.

Purpose:

In this study, monthly and bi-weekly habitual CL wearers were refitted in a water gradient DD SiHy lens (delefilcon A) for four weeks of wear.
Primary and secondary study objectives were the change in bulbar redness and limbal redness after refit, respectively. The exploratory endpoint related to subjective evaluation was wearing comfort.

Methods:

50 of 52 habitual, asymptomatic, full-time hydrogel lens and silicone hydrogel lens wearers (ratio 1:1) with spherical correction completed a prospective study conducted at two sites in Germany. All lens wearers used all-in-one lens care solutions (not Polyquad-based or peroxide solutions).

Baseline measures included objective bulbar and limbal redness using the Oculus K5M Redness-Scan, and subjective comfort ratings and ocular surface staining grades. Comfort was assessed post-insertion and diurnally using a 101-point visual analog scale. Staining was assessed using the 0-4-point JENVIS Grading Scale.

Participants underwent a wash-out phase of 6 ± 1 days of no lens wear before delefilcon A lenses were dispensed (at V2) for a wearing period of 26 ± 5 days. A follow-up visit (V3) was scheduled 6 ± 1 days after V2 and a final visit (V4) after an additional 20 ± 4 days. All measures were repeated at visits 2, 3 and 4. Redness and staining values for the right eye only were considered for analysis. Wilcoxon signed-rank test and t-test for paired samples were used for the statistical analyses.

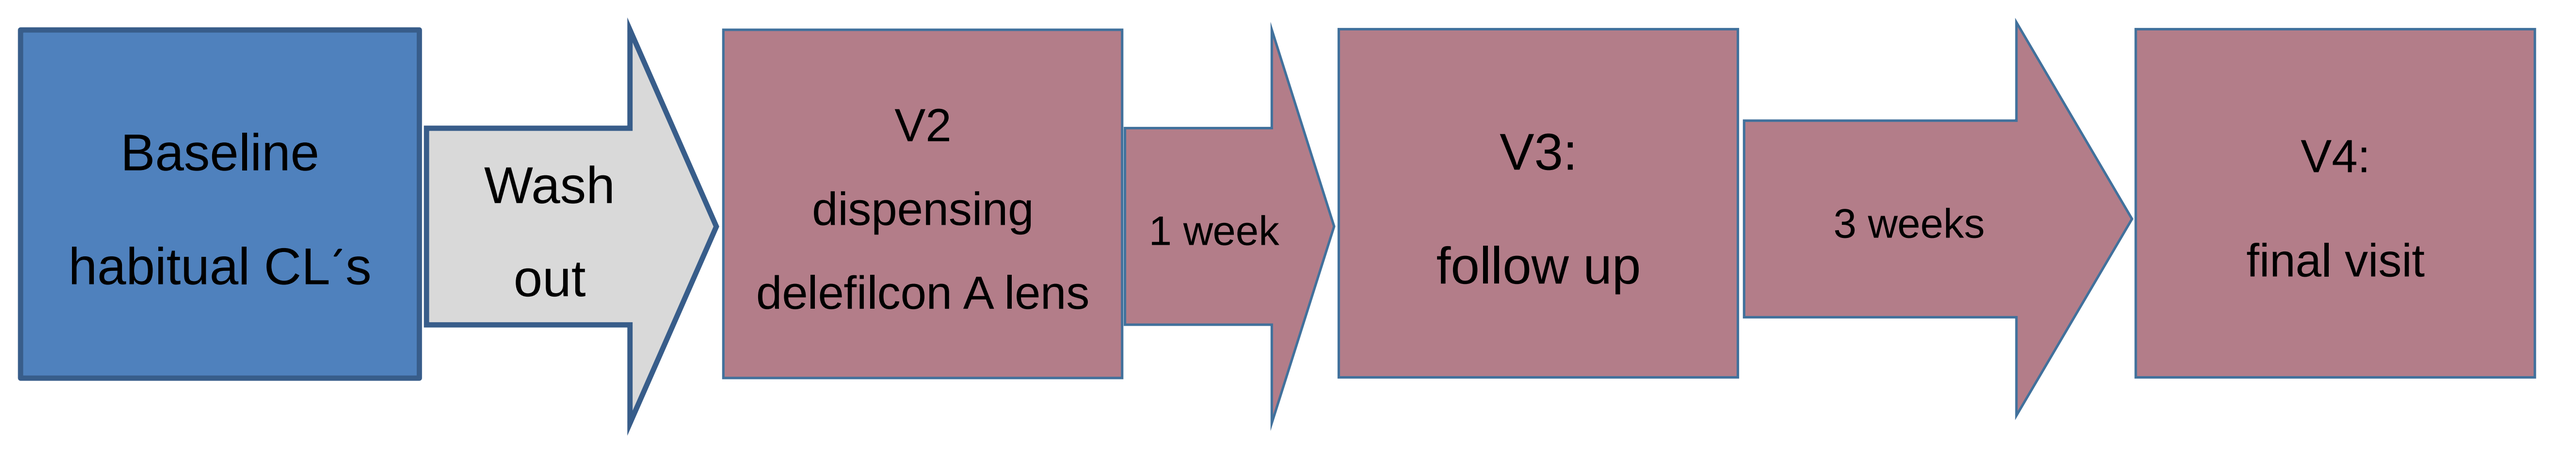
Baseline
habitual CL´s
Wash out
V2
dispensing
delefilcon A lens
1 week
V3:
follow up
3 weeks
V4:
final visit

Figure 1: Visit schedule

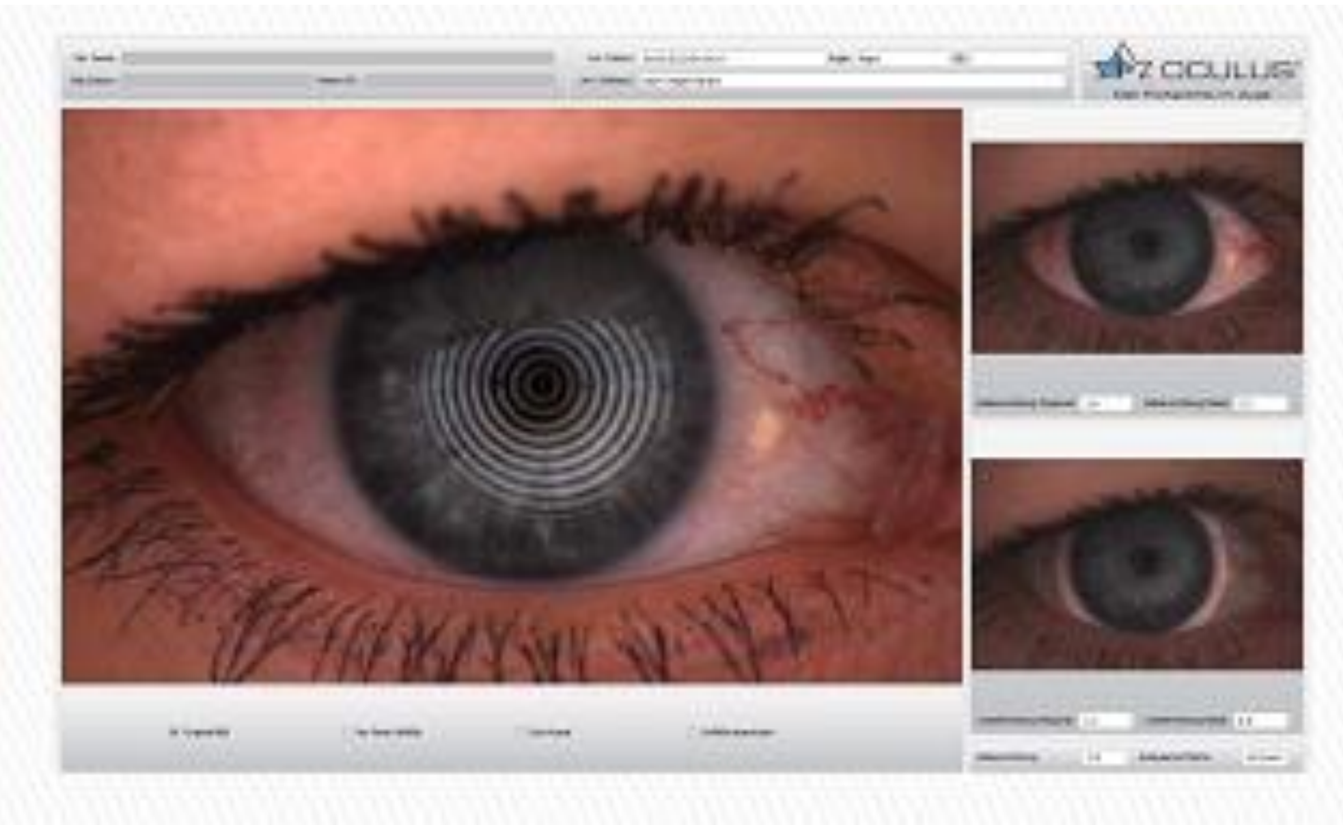

Figure 2: Captured images of inter-palpebral lid aperture were automatically analyzed for bulbar and limbal redness by the R-Scan software module (Oculus Optikgeräte GmbH, Wetzlar, Germany)

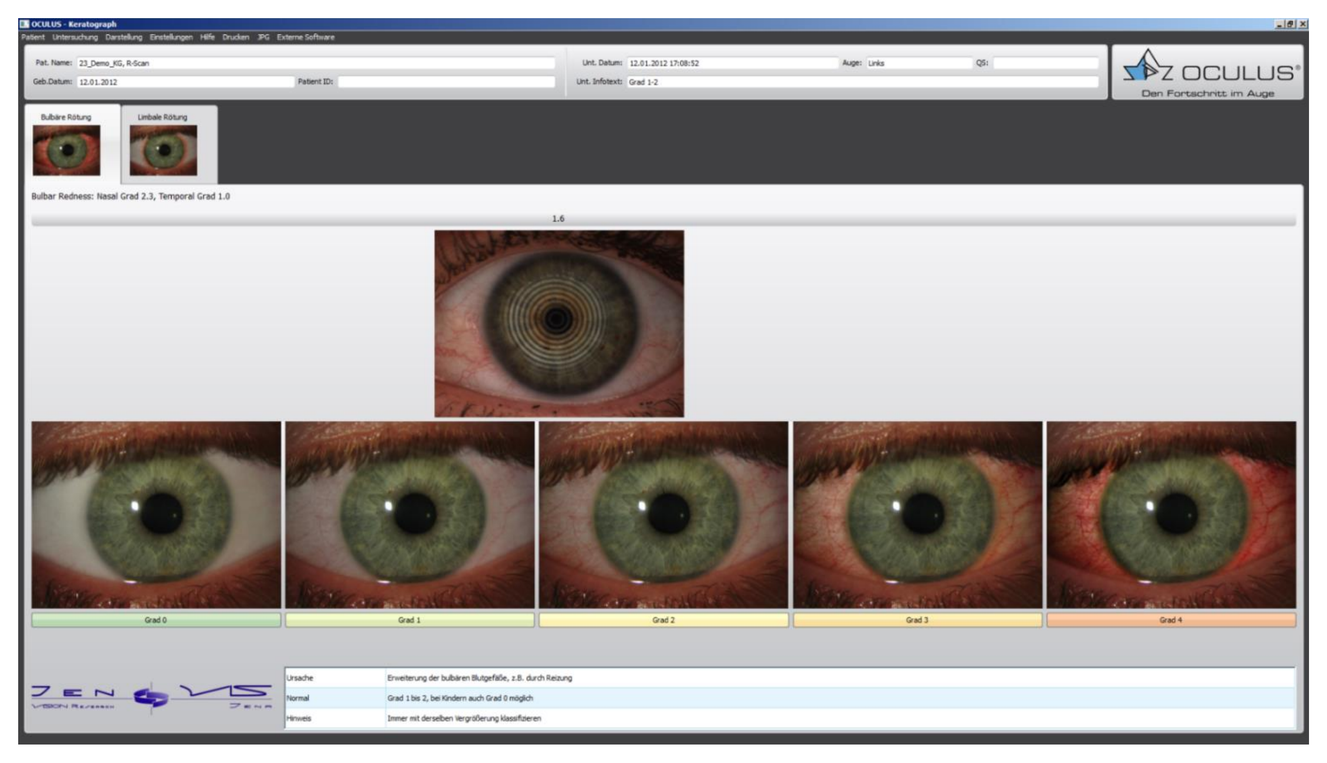

Figure 3: automatic grading is based on the validated grading scale of JENVIS Research [3]

Results:

Bulbar redness decreased in both subgroups over time. The mean score decreased from 0.88 to 0.77 (paired sample test p = 0.012) for previous habitual hydrogel wearers and from 0.83 to 0.71 (paired sample test p = 0.058) for previous silicone hydrogel 2/4w wearers (Figure 4).

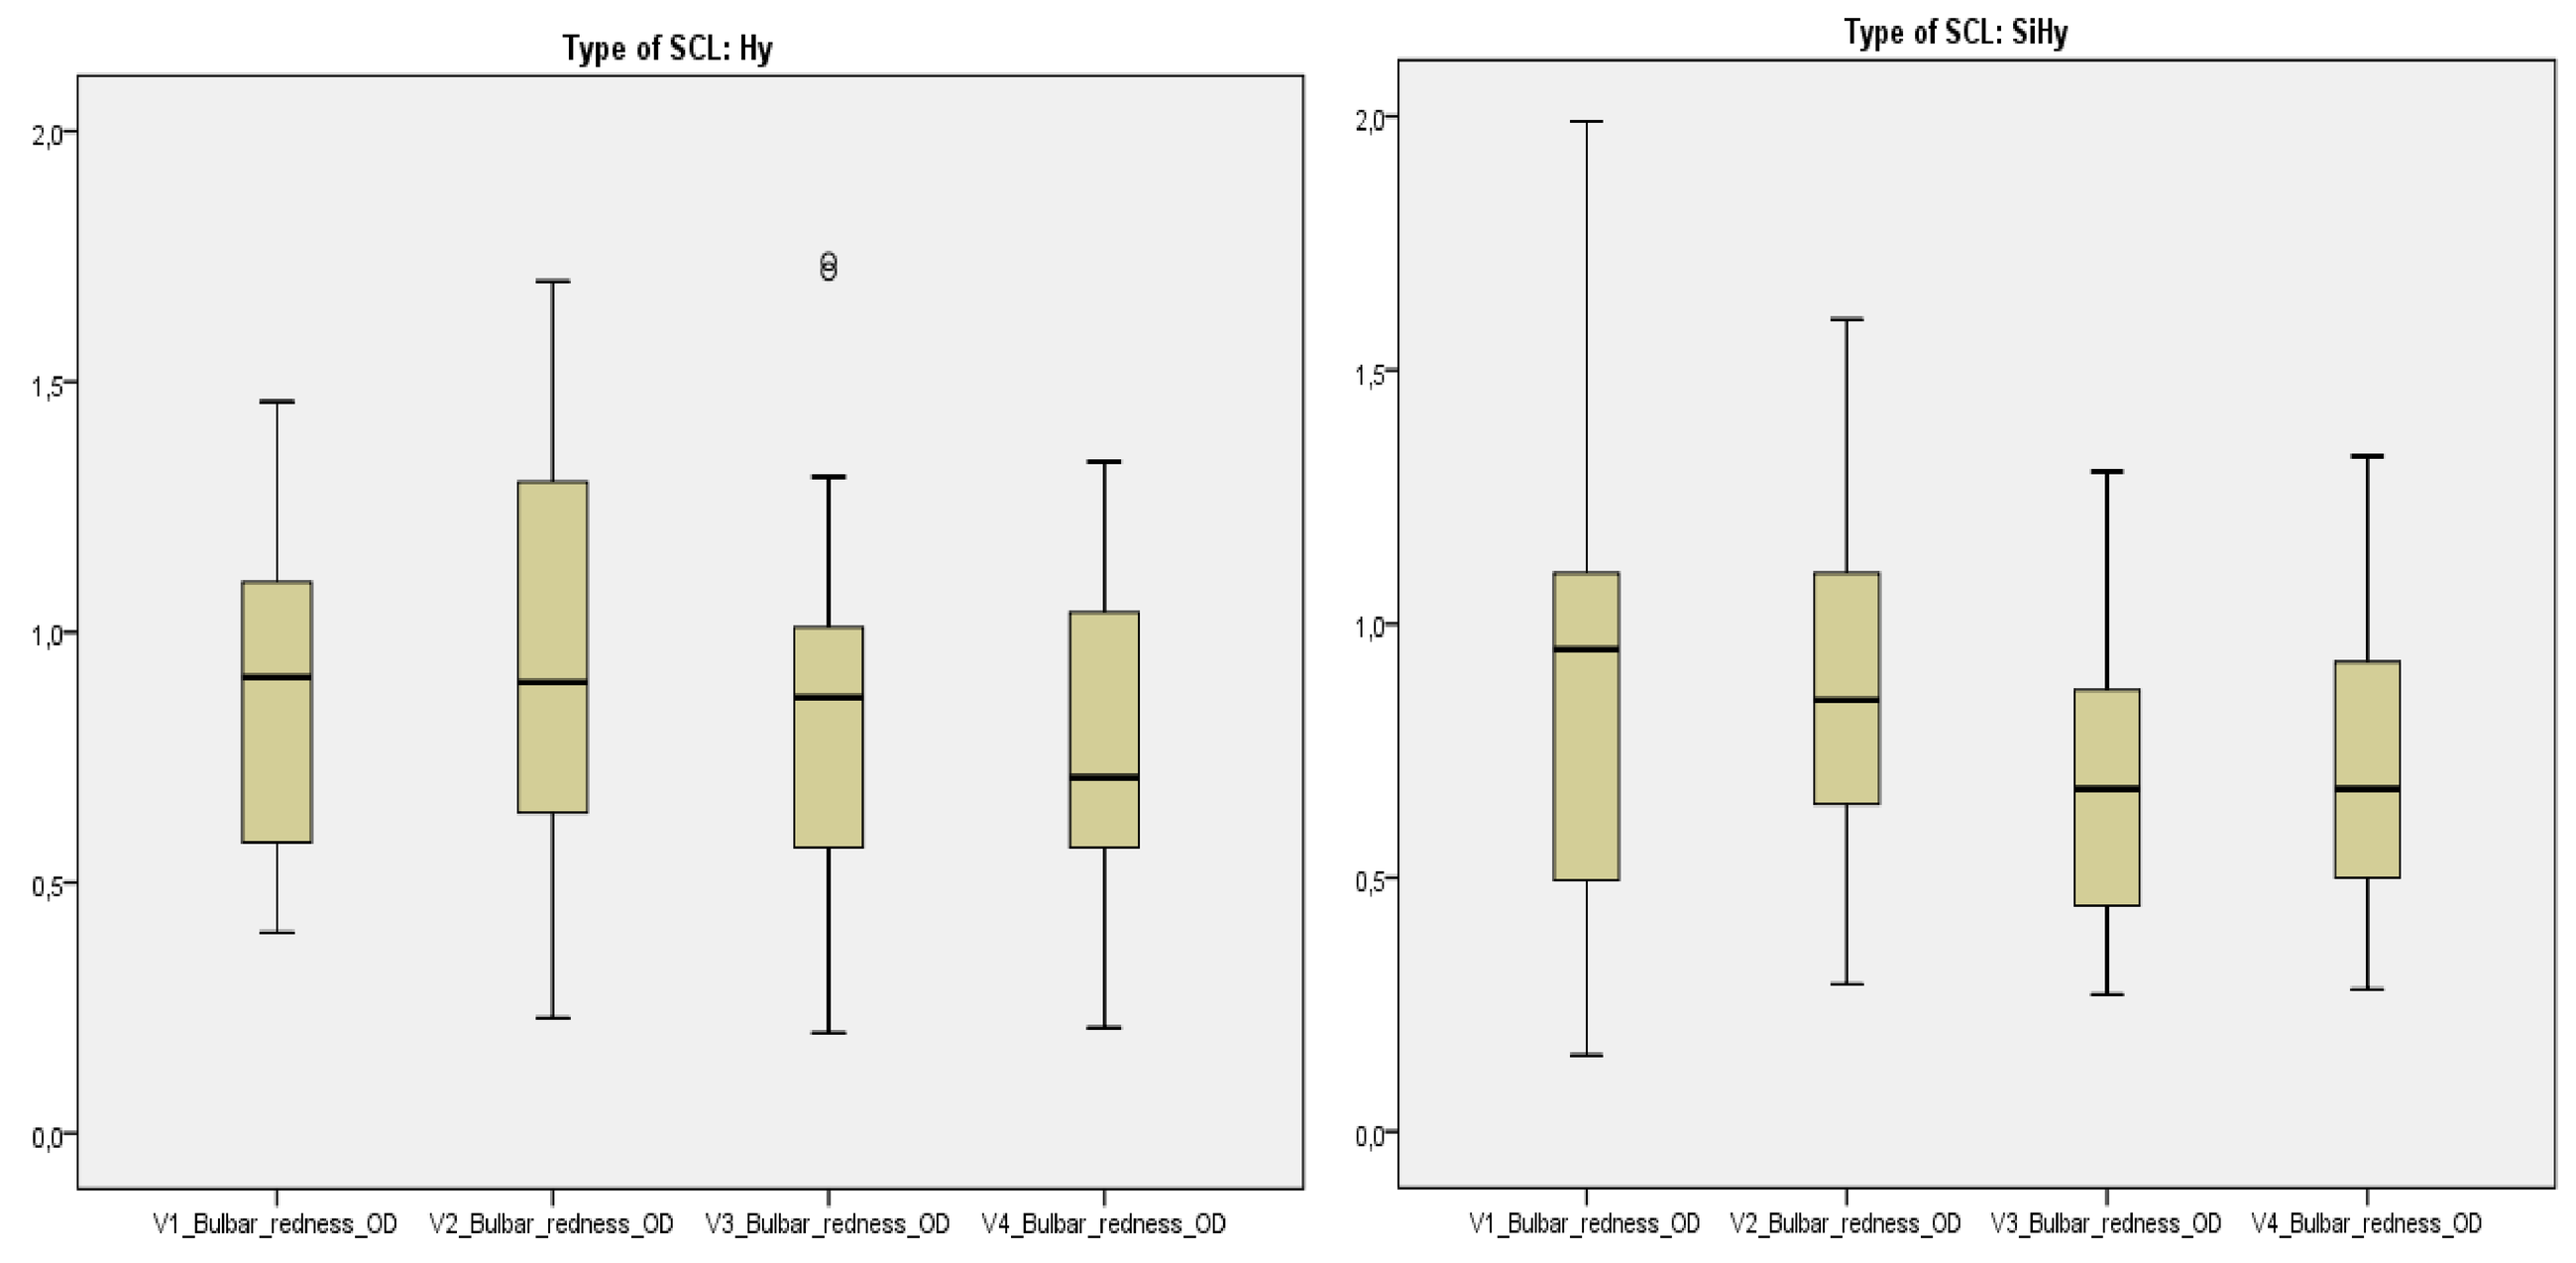
Type of SCL: Hy
V1_Bulbar_redness_OD
V2_Bulbar_redness_OD
V3_Bulbar_redness_OD
V4_Bulbar_redness_OD
Type of SCL: SiHy

Figure 4: Bulbar redness for the hydrogel subgroup (left) and the silicone hydrogel subgroup (right)

Limbal redness decreased in both subgroups over time as well. The mean grade decreased from 0.41 to 0.23 (paired sample test $p < 0.001$) for previous habitual hydrogel wearers and from 0.43 to 0.30 (paired sample test p = 0.052) for previous silicone hydrogel 2/4w wearers (Figure 5).

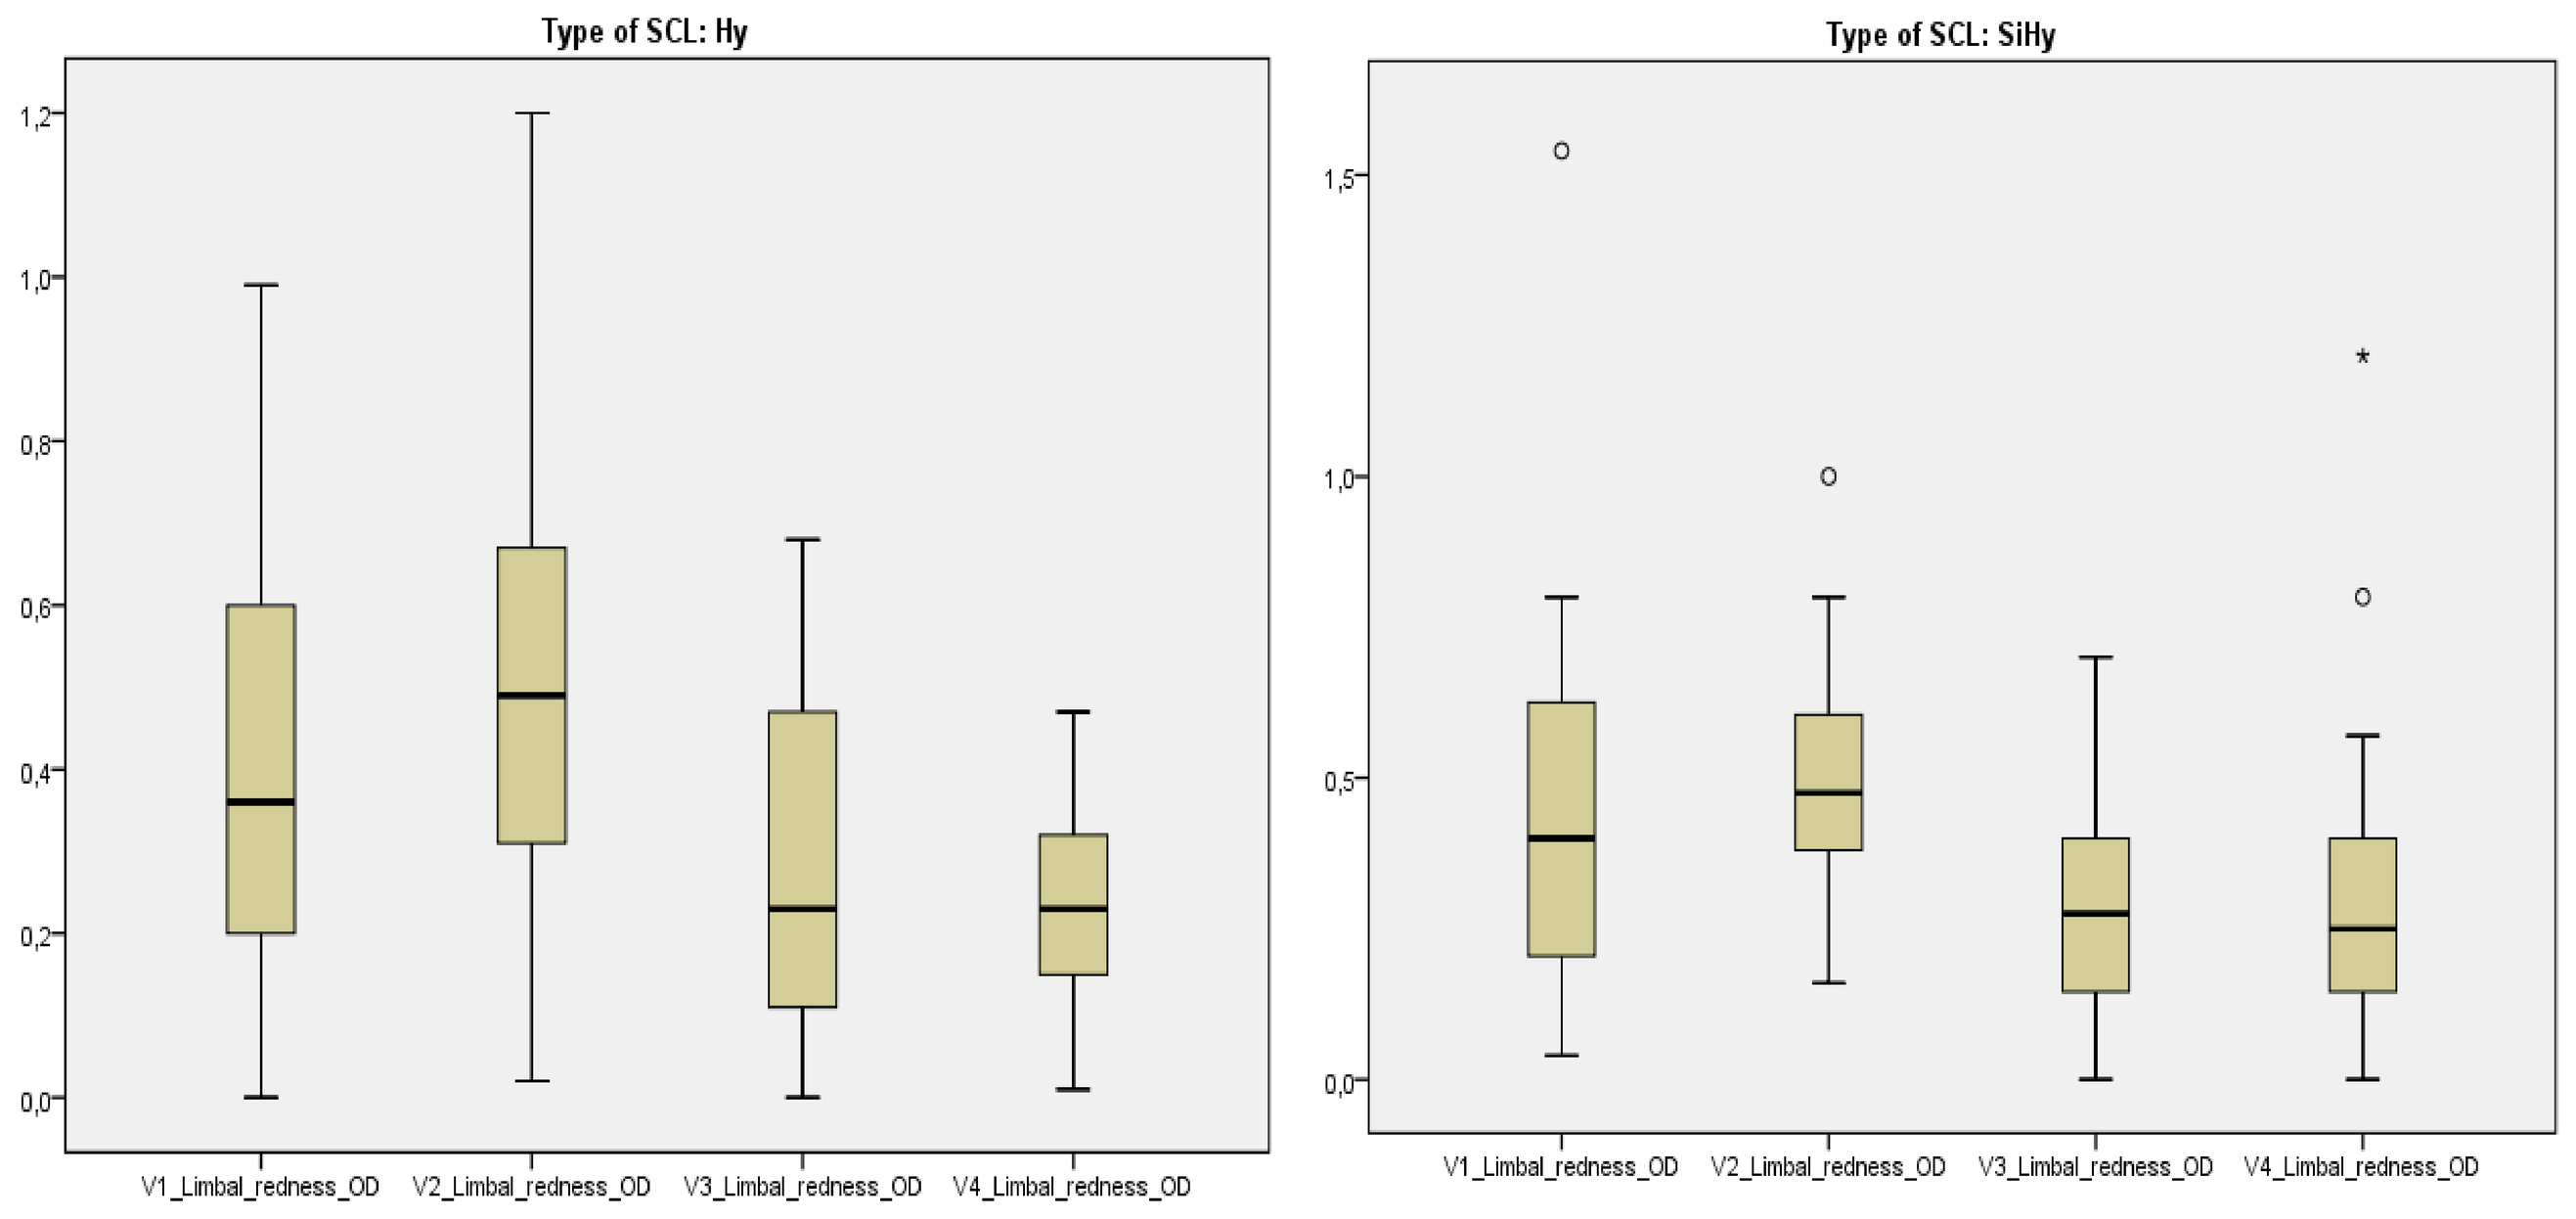
Type of SCL: Hy
V1_Limbal_redness_OD
V2_Limbal_redness_OD
V3_Limbal_redness_OD
V4_Limbal_redness_OD
Type of SCL: SiHy

Figure 5: Limbal redness for the hydrogel subgroup (left) and the silicone hydrogel subgroup (right)

The mean corneal staining grade decreased from 0.28 to 0.04 (Wilcoxon paired rank test p = 0.014) for previous habitual hydrogel wearers and from 0.24 to 0.04 (p = 0.025) for previous silicone hydrogel 2/4w wearers.
The mean bulbar staining grade decreased from 0.28 to 0.12 (Wilcoxon paired rank test p = 0.046) for previous habitual hydrogel wearers and from 0.56 to 0.32 (p = 0.166) for previous silicone hydrogel 2/4w wearers.
Subjectively evaluated comfort was rated higher in both subgroups with the daily disposable lens in comparison to the habitually used 2-4 week contact lens (Wilcoxon paired rank test p= 0.003 for hydrogel and p = 0.009 for SiHy subgroup.

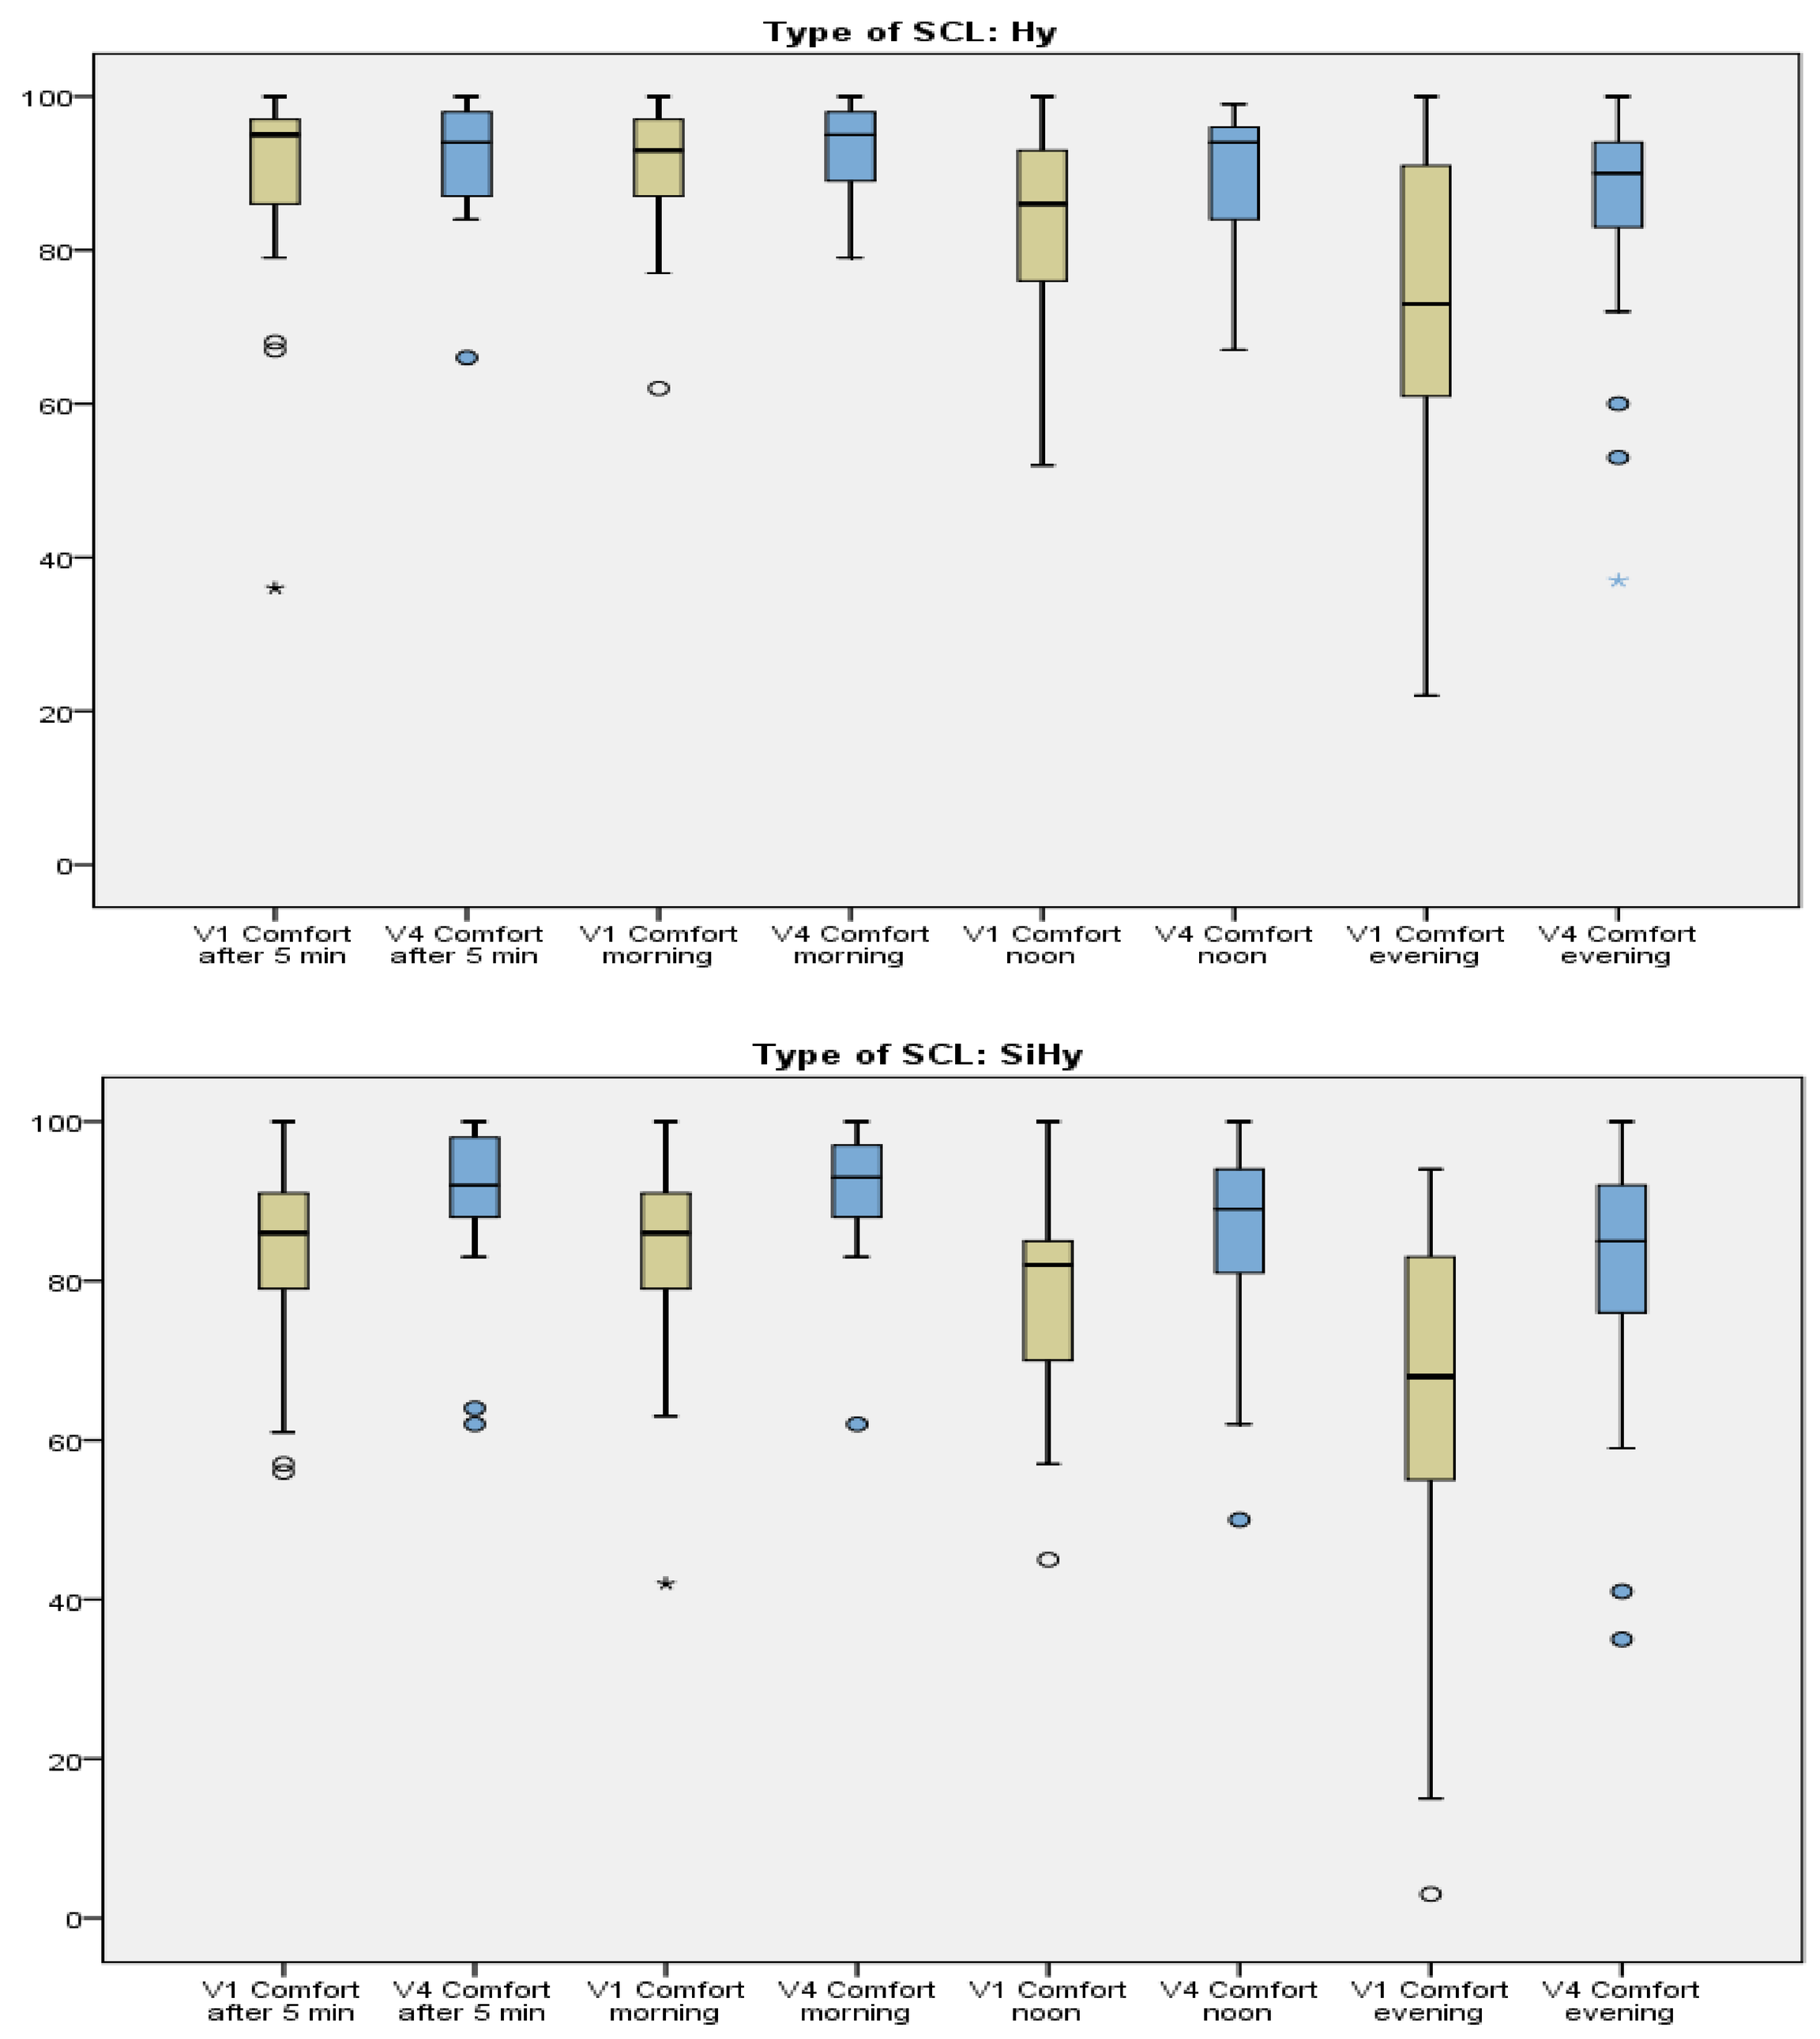
Type of SCL: Hy
V1 Comfort after 5 min
V4 Comfort after 5 min
V1 Comfort morning
V4 Comfort morning
V1 Comfort noon
V4 Comfort noon
V1 Comfort evening
V4 Comfort evening
Type of SCL: SiHy

Figure 6 and 7: Subjective comfort for the hydrogel subgroup (top) and the silicone hydrogel subgroup (bottom) at visit 1 and visit 4

Conclusion:

Although differences were small in the context of classic subjective grading scales, which use full grades in 1.0 steps, the Oculus K5M was able to detect fine graduated differences in a cohort of non-symptomatic wearers presenting with mild redness. The refit to a daily disposable delefilcon A lens after a wash-out phase was beneficial, with reduced bulbar and limbal redness, reduced corneal and conjunctival staining and increased wearer comfort. These benefits were slightly higher for the habitual hydrogel lens wearer subgroup.

References:

1. Papas E: The role of hypoxia in the limbal vascular response to soft contact lens wear; Eye Contact Lens. 2003 Jan;29(1 Suppl):S72-4; discussion S83-4, S192-4.
2. Muntz A, Subbaraman L, Sorbara L, Jones L: Tear exchange and contact lenses: a review, J Optom Jan-Mar 2015;8(1):2-11.doi: 10.1016/j.optom.2014.12.001. Epub 2015 Jan 7.
3. Sickenberger W, Oehring D: Validation of a novel morphing software to classify different slit lamp findings; Contact Lens & Anterior Eye 35(1), DOI: 10.1016/j.clae.2012.08.065

Acknowledgements:

The IIT study # 43410197was fully funded by Alcon Inc..

contact: jenvis-research.com | Address: JENVIS Research Institute • c/o Ernst Abbe University of Applied Sciences Jena • Carl-Zeiss-Promenade 2 • 07745 Jena • Germany | Phone: 0049-(0)3641-205871 | E-Mail: s.marx@jenvis-research.com